Poster number
19T0900

DFE pharma

2016 AAPS
ANNUAL MEETING AND EXPOSITION
NOVEMBER 13–17, 2016
COLORADO CONVENTION CENTER, DENVER
aaps®

Timo Schigt and Bastiaan HJ Dickhoff
DFE Pharma, Kleverstrasse 187, 47574 Goch, Germany

Continuous improvement of highly consistent excipient manufacturing using QbD principles – An excipient manufacturer's perspective

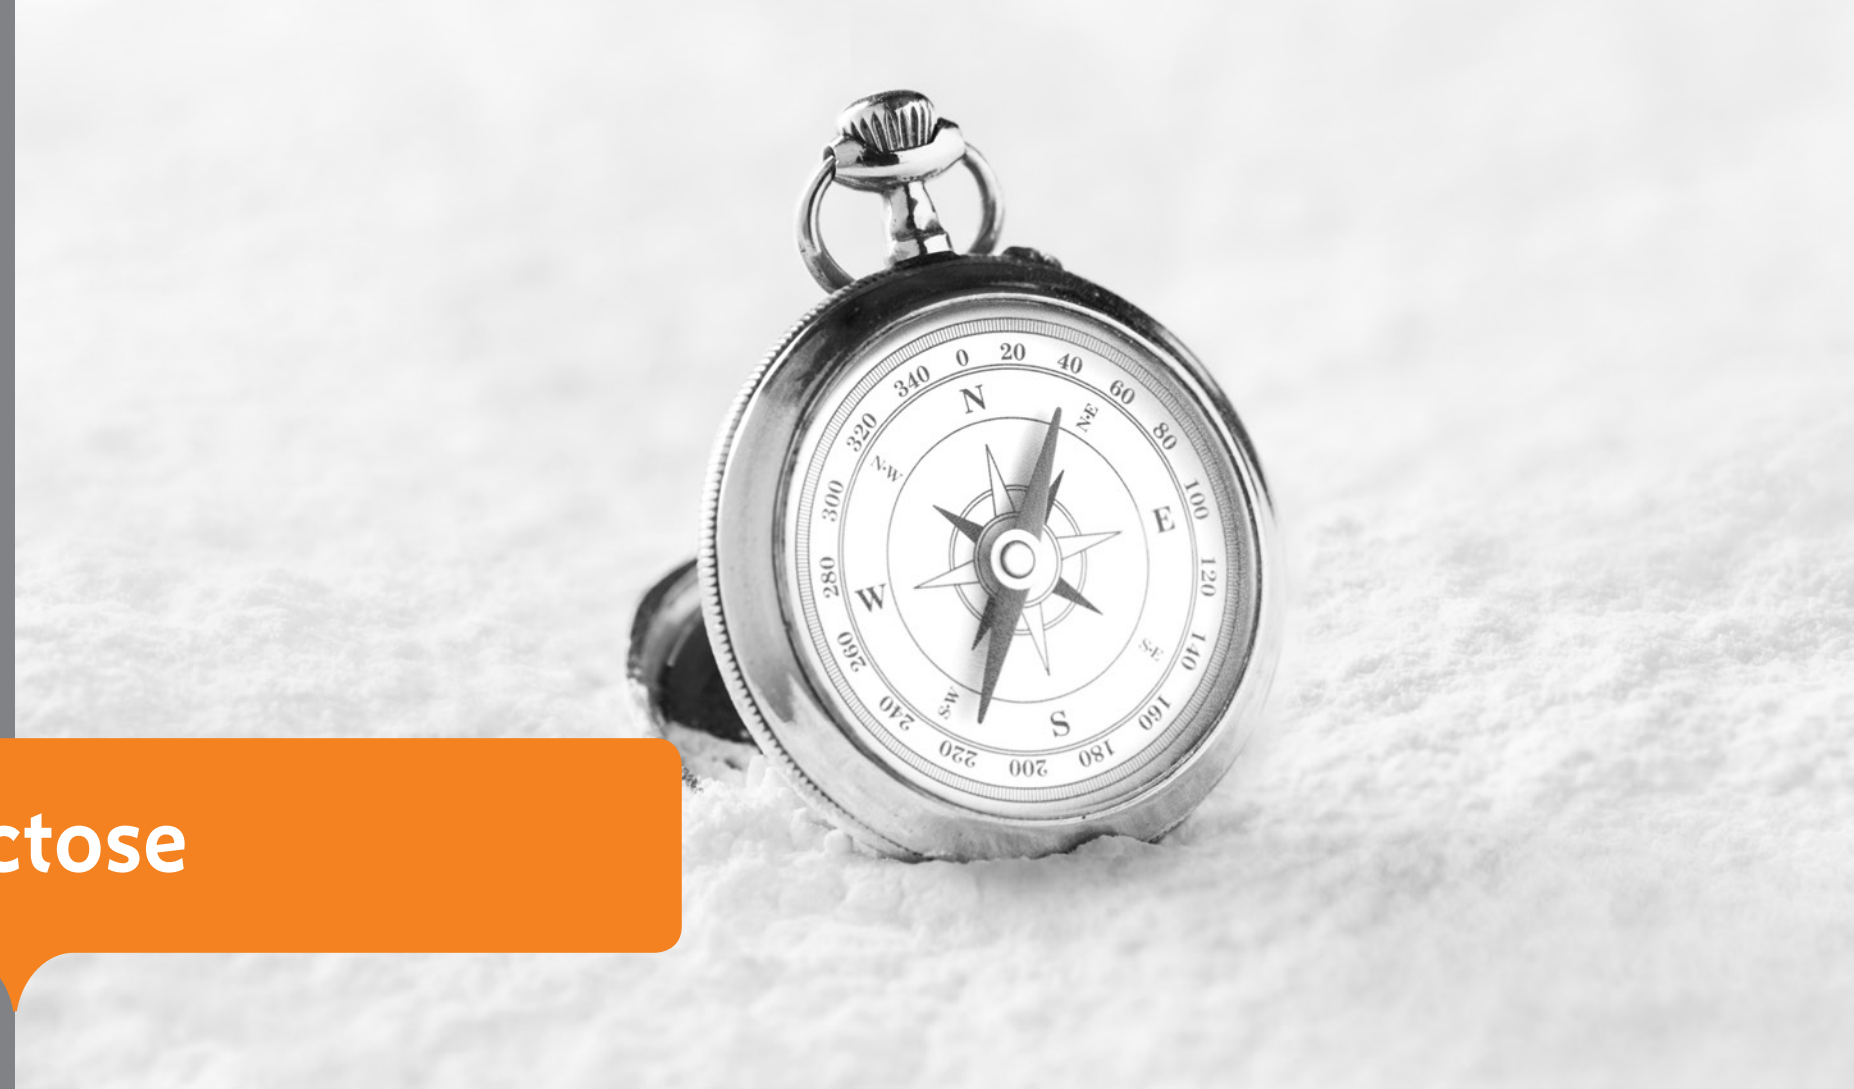

Lactose

Purpose

The Quality by Design (QbD) approach is defined by the ICH Q8 guideline and titled the "quality of the 21st century" by the FDA in 2003. QbD requires understanding of the product critical quality attributes (CQAs) to translate these in a design space for the process. The design space sets target ranges for the critical process parameters (CPPs) based on the likelihood that the resulting product will meet the CQA specifications.

Principles of QbD are applicable in existing products and processes as well. An existing product has a set of historical data which can be used as an extensive DOE for the examination of the design space, limited by the variation that has actually occurred in production. The new information in the design space can be used to narrow the CPPs target ranges where appropriate to maximize the percentage first time right and increase product consistency.

Main objective of this work is to present an excipient manufacturers view on how to use principles of QbD in the continuous improvement of existing excipient products throughout the production chain, to further increase product consistency.

Methods

SuperTab® 11SD (EU) is used as an example to illustrate our proposed approach. SuperTab® 11SD is a functional spray-dried lactose grade commonly used in direct compression. SuperTab® 11SD is manufactured in a semi-continuous process focused on high product quality and consistency. Data from throughout the manufacturing process for a period of five years of manufacturing were taken into account.

Multivariate mathematical method principal component analysis (PCA) is ideally suited to analyze big data sets. It transforms the high dimensional data into a lower dimensional set of principal components (PCs). The method gains a risk-based character if only the first and most important principal components are further analyzed.

Results

The PCA model on SuperTab® 11SD end product quality parameters (Figure 1) shows very little variation explained on the first two components (17 and 11.2% respectively), indicating a low amount of structured variation. The batches produced in the year 2014 show a slight drift (well within product- and pharmacopeial specifications) to the right along the first PC. Examination of the PCA model on the process parameters of selected batches in the period 2013-2014 (Figure 2) shows a shift in the process parameters. Understanding the shift in process parameters and their impact on product properties enabled to focus on a risk based process control. The involved loadings of both models could be related and were used to validate the CPPs as defined based on process experience. This enabled a more consistent product in 2015-2016 (not shown).

Conclusion

Principal component analysis of data throughout the production chain of an existing excipient product has shown the power of multivariate process control and the ability to challenge and set the process CPPs. The use of QbD principles will further increase excipient consistency, de-risking their use in pharmaceutical formulations.

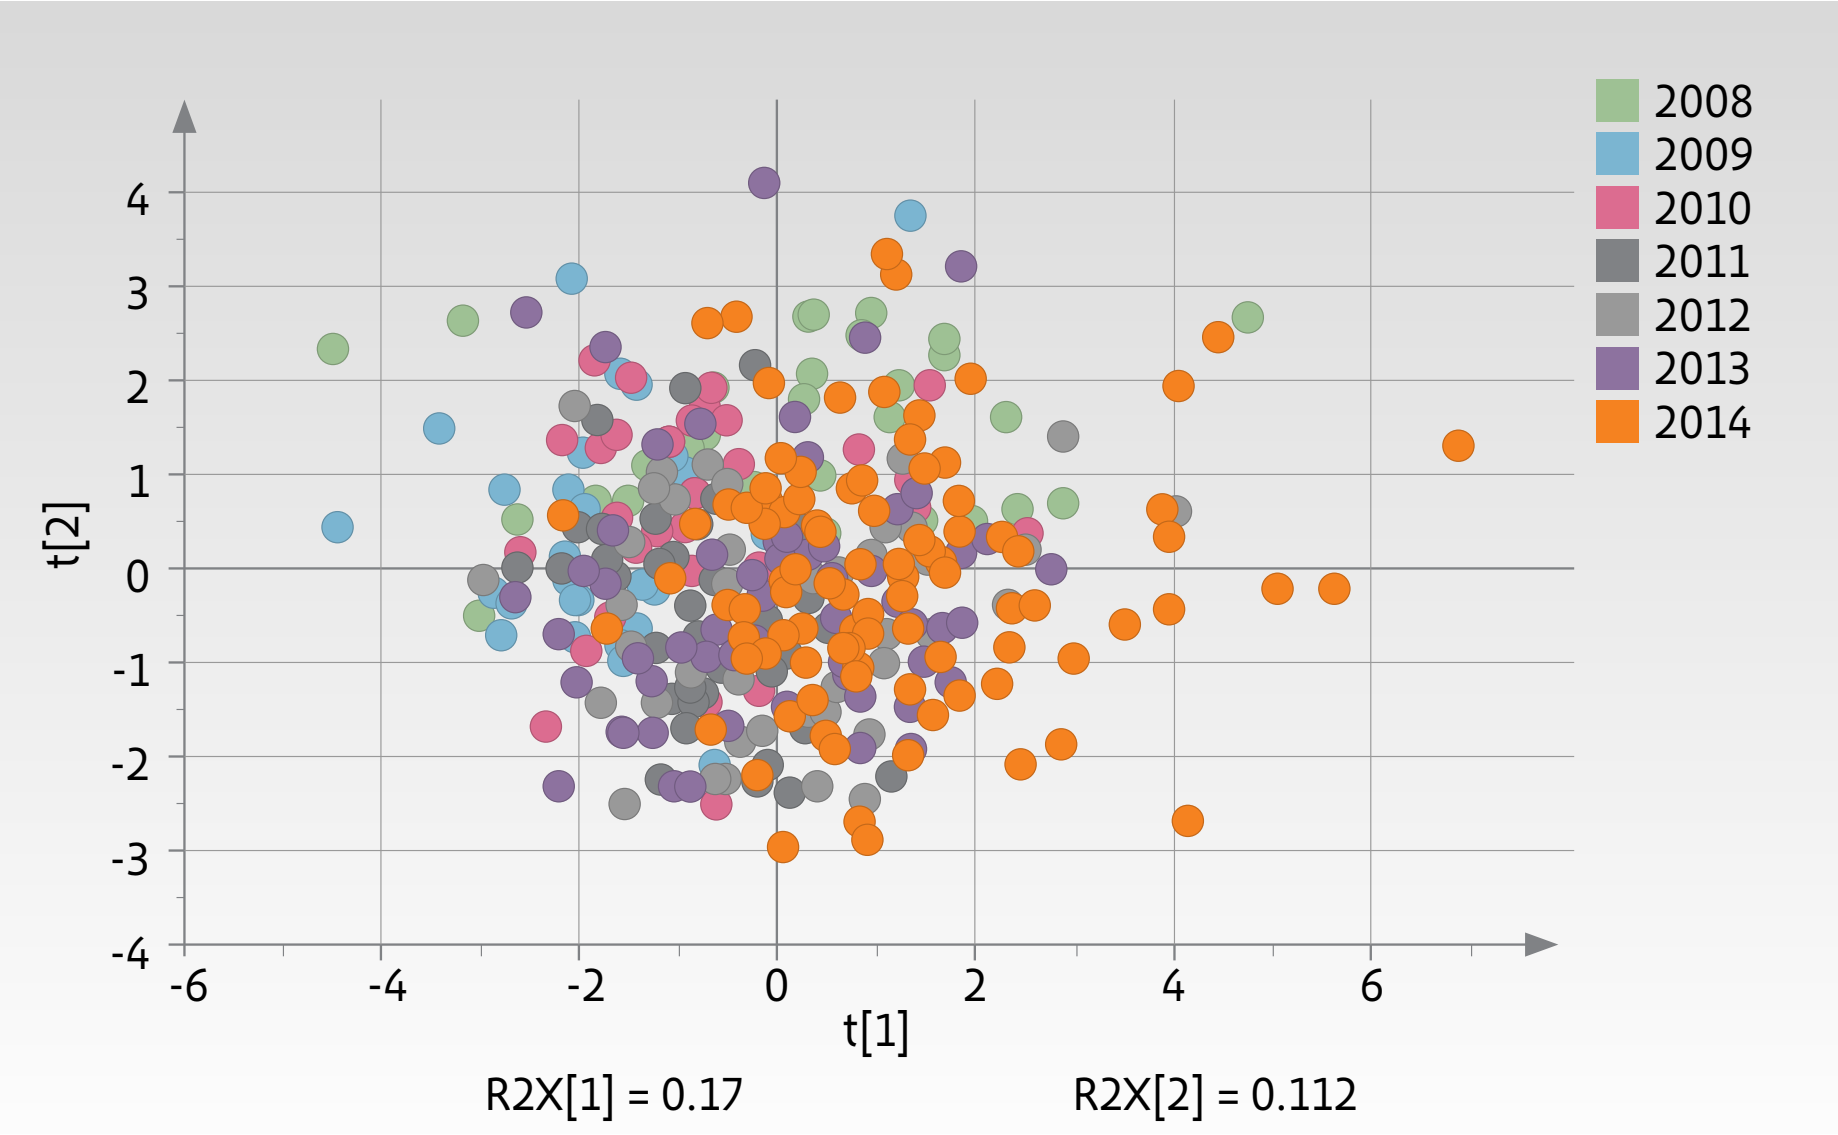
2008
2009
2010
2011
2012
2013
2014
t[2]
t[1]
R2X[1] = 0.17
R2X[2] = 0.112

Figure 1: *Score plot of the PCA model on end product quality parameters of released SuperTab® 11SD batches produced between 2008 and 2014, colored by the year of production.*

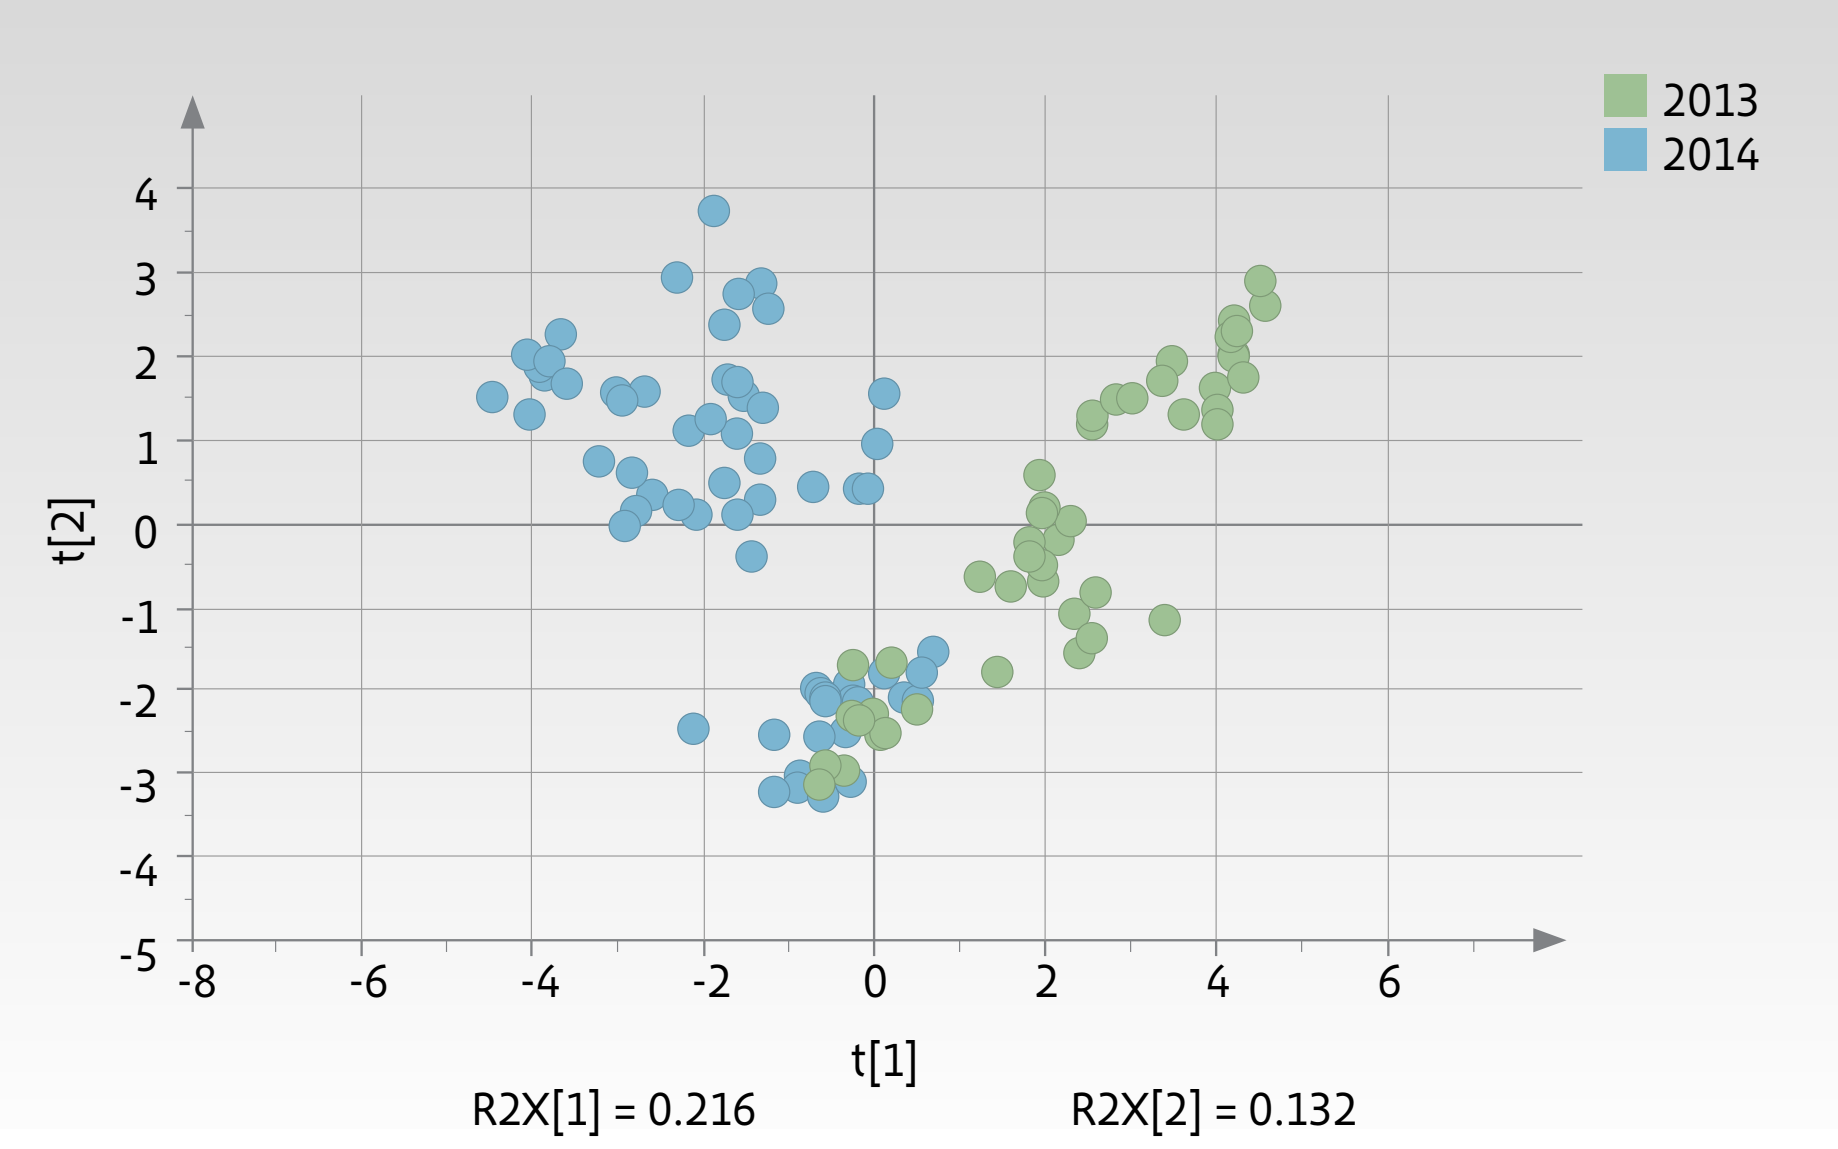
2013
2014
t[2]
t[1]
R2X[1] = 0.216
R2X[2] = 0.132

Figure 2: *Score plot of the PCA model on process parameters of batches SuperTab 11SD produced in the period 2013-2014, colored by the year of production.*

www.dfepharma.com

The pursuit of excipient excellence